Healthcare Resource Utilization Following 6 Months of Treatment With Olanzapine/Samidorphan: Real-World Assessment of Patients With Schizophrenia or Bipolar I Disorder

Andrew Cutler,[1,2] Hemangi R. Panchmatia,[3] Alejandro G. Hughes,[4] Noah S. Webb,[4] Michael J. Doane,[3] Rakesh Jain[5]

[1]SUNY Upstate Medical University, Syracuse, NY; [2]Neuroscience Education Institute, Lakewood Ranch, FL; [3]Alkermes, Inc., Waltham, MA; [4]Optum, Inc., Eden Prairie, MN; [5]Department of Psychiatry, Texas Tech University School of Medicine-Permian Basin, Midland, TX

INTRODUCTION

- Long-term pharmacotherapy with antipsychotic medication is recommended for treating schizophrenia (SZ) and bipolar I disorder (BD-I)[1,2]
- However, adverse effects of atypical antipsychotics, including weight gain, contribute to nonadherence and treatment discontinuation,[3] potentially leading to hospitalization and other healthcare resource utilization (HCRU)[4,5]
- The combination of olanzapine and samidorphan (OLZ/SAM) provides the established antipsychotic efficacy of olanzapine[6] while mitigating olanzapine-associated weight gain[7]; several clinical trials have demonstrated these effects[8-10]
- Until now, no real-world studies have examined HCRU among patients with SZ or BD-I who have initiated treatment with OLZ/SAM
- This analysis aimed to understand if OLZ/SAM may be associated with reductions in HCRU in patients in a real-world setting
- The objective was to assess and compare HCRU 6 months before and after initiating OLZ/SAM in patients treated for SZ or BD-I

METHODS

Data Source

- This retrospective study used administrative claims data from April 19, 2021, through December 31, 2022 (study period), obtained from Komodo Healthcare Map, a fully deidentified US-based database containing detailed information on inpatient (IP), outpatient (OP), and pharmacy claims data from ~150 million patients covered by commercial (66%), Medicaid (27%), or Medicare Advantage (7%) plans

Patients and Study Design

Inclusion Criteria

- Adults (aged ≥18 years) with ≥1 medical claim for SZ or BD-I (**Figure 1** and **Figure 2**)
- ≥1 pharmacy or medical claim for OLZ/SAM
- ≥6 months of continuous enrollment with medical and pharmacy benefits before (baseline period) and after (fixed follow-up period) the index date (date of first medical or pharmacy claim for OLZ/SAM)
- ≥1 medical claim for SZ or BD-I during the baseline or follow-up period
 - Patients with medical claims for both SZ and BD-I were assigned an indication of SZ

Exclusion Criteria

- Patients with pharmacy or medical claims for OLZ/SAM during the baseline period

Figure 1. Study Design

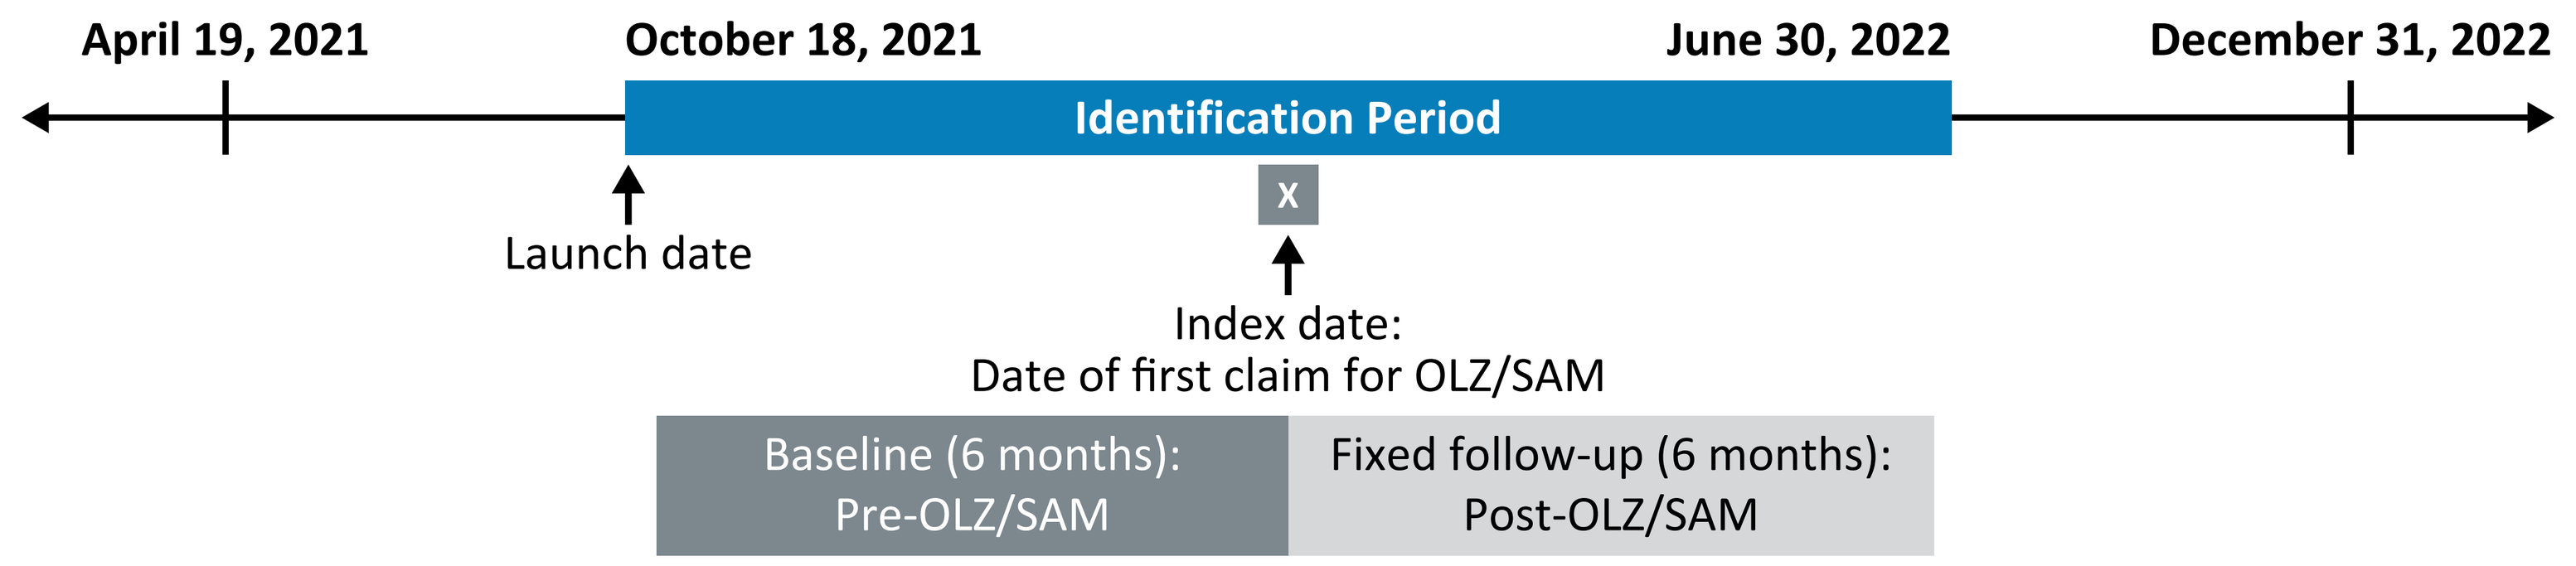
April 19, 2021
October 18, 2021
June 30, 2022
December 31, 2022
Identification Period
Launch date
X
Index date:
Date of first claim for OLZ/SAM
Baseline (6 months):
Pre-OLZ/SAM
Fixed follow-up (6 months):
Post-OLZ/SAM

OLZ/SAM, combination olanzapine and samidorphan.

Outcomes

- Treatment patterns, including discontinuation, adherence, and persistence
- HCRU, including IP admissions, emergency department (ED) and OP visits, and average number of days in hospital per patient in each of the following categories:
 - All-cause HCRU
 - Mental health–related HCRU
- Baseline demographics, clinical characteristics, and medication use

Statistics

- For baseline patient demographics and clinical characteristics, numbers and percentages were reported for categorical variables, and means and SDs were reported for continuous variables
- Six-month pre-post comparisons (baseline vs follow-up) were made using the following unadjusted pairwise comparisons:
 - Paired t-test for normally distributed continuous variables
 - Wilcoxon signed-rank test for distribution-free continuous variables
 - McNemar test for dichotomous variables
- A completer analysis of HCRU was conducted in the subset of patients who completed the full 6 months of follow-up

RESULTS

Patients

- The analysis included data from 855 patients with SZ and 691 with BD-I (**Figure 2**; **Table 1** and **Table 2**)

Figure 2. Patient Identification

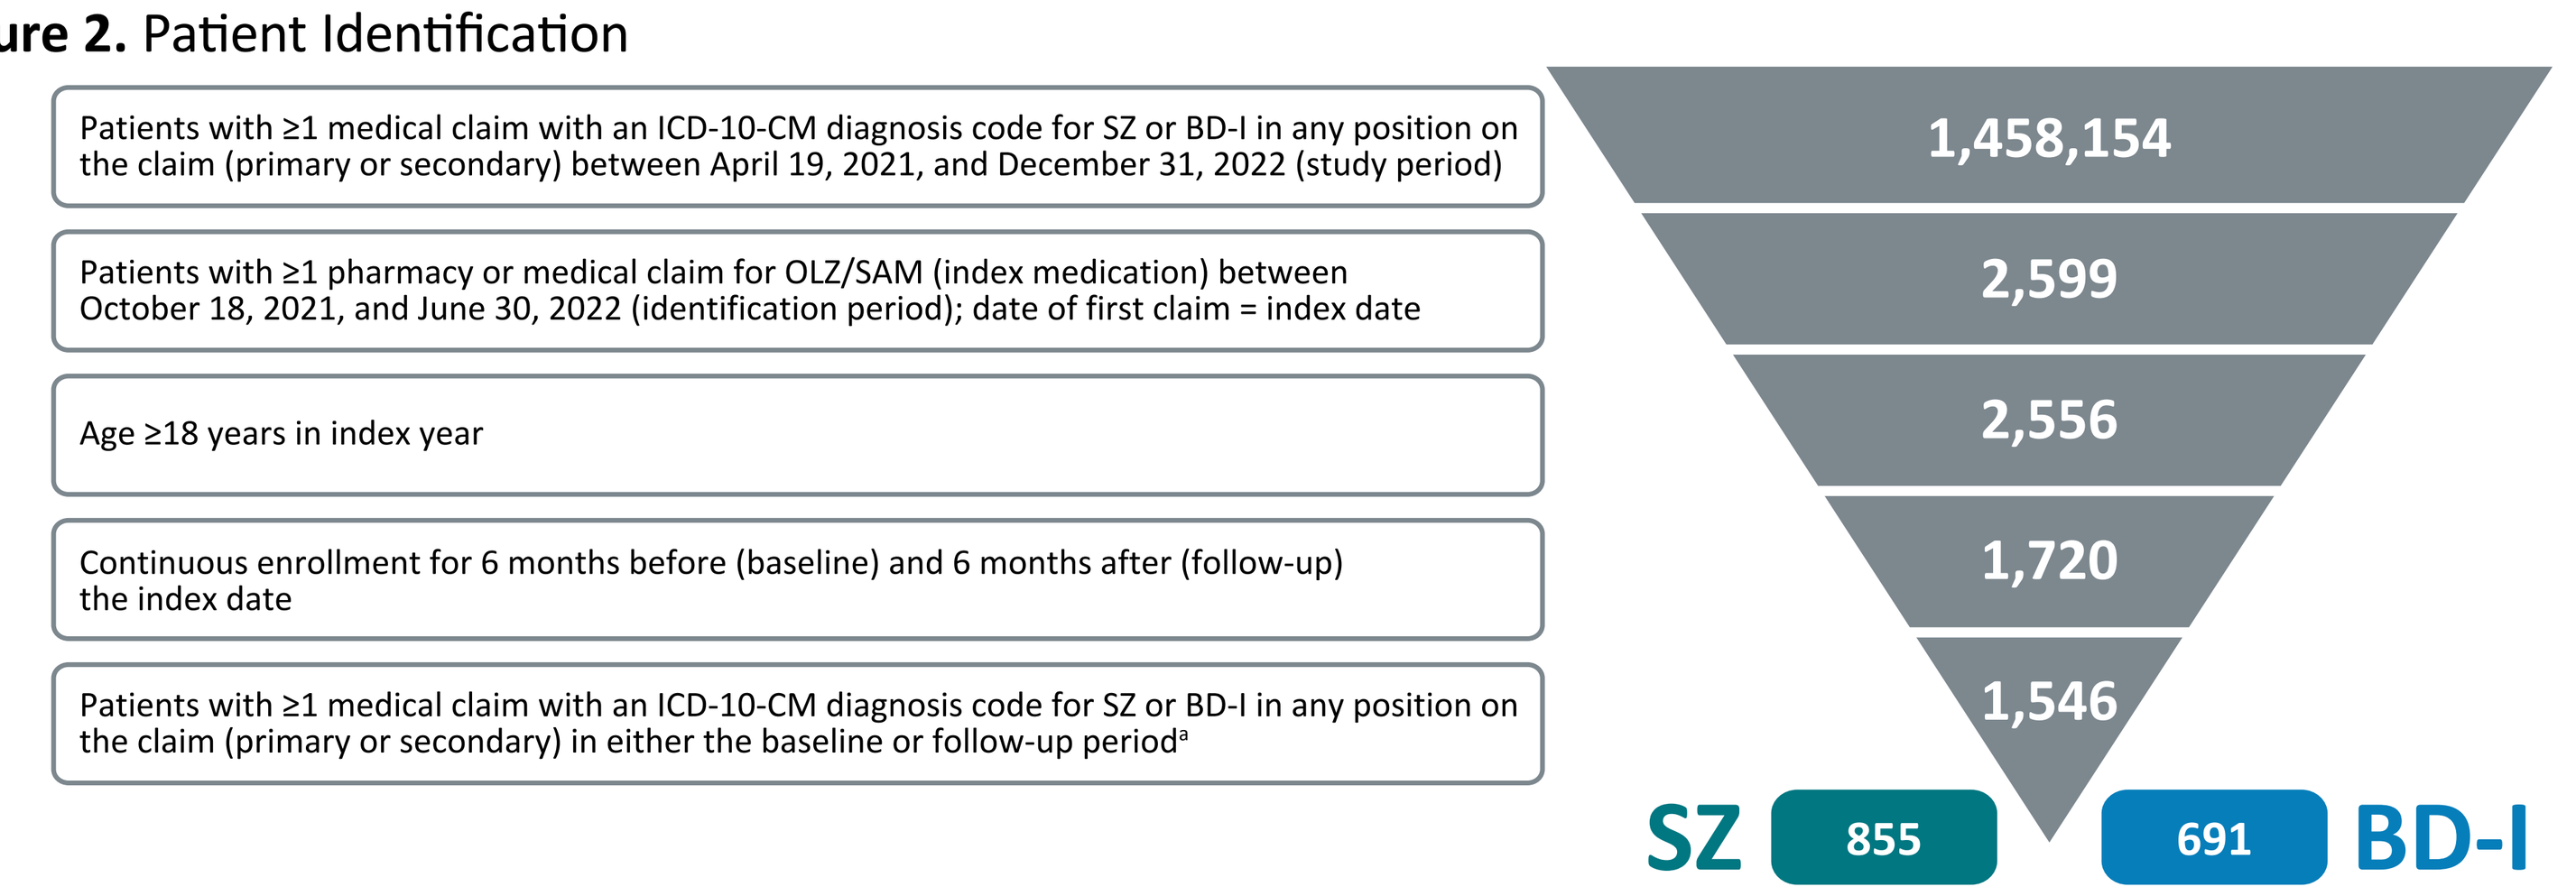
Patients with ≥1 medical claim with an ICD-10-CM diagnosis code for SZ or BD-I in any position on the claim (primary or secondary) between April 19, 2021, and December 31, 2022 (study period)
Patients with ≥1 pharmacy or medical claim for OLZ/SAM (index medication) between October 18, 2021, and June 30, 2022 (identification period); date of first claim = index date
Age ≥18 years in index year
Continuous enrollment for 6 months before (baseline) and 6 months after (follow-up) the index date
Patients with ≥1 medical claim with an ICD-10-CM diagnosis code for SZ or BD-I in any position on the claim (primary or secondary) in either the baseline or follow-up period[a]
1,458,154
2,599
2,556
1,720
1,546
SZ
855
691
BD-I

[a]Patients with medical claims for both SZ and BD-I were assigned an indication of SZ (n=129); patients with pharmacy or medical claims for the index medication during the baseline period were excluded (n=2).
BD-I, bipolar I disorder; ICD-10-CM, *International Classification of Diseases, 10th Revision, Clinical Modification*; OLZ/SAM, combination olanzapine and samidorphan; SZ, schizophrenia.

Table 1. Baseline Patient Demographics

Characteristics	SZ Cohort (n=855)	BD-I Cohort (n=691)
Age, mean (SD), years	39.4 (13.6)	38.4 (12.0)
Sex, n (%)		
Female	405 (47.4)	477 (69.0)
Male	450 (52.6)	214 (31.0)
Region, n (%)		
South	243 (28.4)	212 (30.7)
Midwest	240 (28.1)	224 (32.4)
West	215 (25.1)	124 (18.0)
Northeast	97 (11.3)	97 (14.0)
Other[a]	60 (7.0)	34 (4.9)
Insurance type, n (%)		
Medicaid	549 (64.2)	354 (51.2)
Medicare	114 (13.3)	48 (7.0)
Multiple	105 (12.3)	73 (10.6)
Commercial	85 (9.9)	213 (30.8)
Unknown	2 (0.2)	3 (0.4)
Index year, n (%)		
2021	200 (23.4)	169 (24.5)
2022	655 (76.6)	522 (75.5)

[a]Other includes Armed Forces Americas (except Canada), Armed Forces (Europe, Canada, Middle East, Africa), Armed Forces Pacific, American Samoa, Federated State of Micronesia, Guam, Marshall Islands, Commonwealth of the Northern Mariana Islands, Puerto Rico, Palau, and Virgin Islands.
BD-I, bipolar I disorder; SZ, schizophrenia.

Table 2. Baseline Clinical Characteristics

Characteristics	SZ Cohort (n=855)	BD-I Cohort (n=691)
Health characteristics reported by ≥10% of patients during baseline period, n (%)		
Anxiety disorders	367 (42.9)	450 (65.1)
Hypertension	284 (33.2)	196 (28.4)
Obesity	262 (30.6)	204 (29.5)
Any substance use disorder	261 (30.5)	223 (32.3)
Major depressive disorder	258 (30.2)	254 (36.8)
Hyperlipidemia	255 (29.8)	167 (24.2)
Type 2 DM	172 (20.1)	96 (13.9)
Posttraumatic stress disorder	134 (15.7)	191 (27.6)
Alcohol use disorder	108 (12.6)	101 (14.6)
Last antipsychotic use before index date, n (%)		
Any second-generation oral[a]	691 (80.8)	578 (83.6)
Any second-generation LAI	70 (8.2)	22 (3.2)
Any first-generation oral	55 (6.4)	19 (2.7)
Any first-generation LAI	15 (1.8)	1 (0.1)
None	47 (5.5)	78 (11.3)
Other common medications taken during baseline period, n (%)		
Antidepressants	553 (64.7)	470 (68.0)
Mood stabilizers	410 (48.0)	490 (70.9)
Antihypertensives	390 (45.6)	321 (46.5)
Anxiolytics	346 (40.5)	356 (51.5)
Metformin, overall	144 (16.8)	97 (14.0)
Metformin, patients with type 2 DM	100 (69.4)	49 (50.5)
Metformin, patients without type 2 DM	44 (30.6)	48 (49.5)

[a]Olanzapine was used by 50% of patients with SZ and 45% of those with BD-I; quetiapine was used by 9% of patients with SZ and 11% of those with BD-I.
BD-I, bipolar I disorder; DM, diabetes mellitus; LAI, long-acting injectable; SZ, schizophrenia.

HCRU in SZ Cohort

- Significant reductions from baseline in the proportions of patients with all-cause (**Figure 3**) and mental health–related (**Figure 4**) IP admissions and ED visits were observed in the 6 months after OLZ/SAM initiation
 - Average all-cause number of days in hospital per SZ patient decreased from 5.9 to 4.8 days (P=0.075)
 - Average mental health–related number of days in hospital per SZ patient decreased from 5.7 to 4.5 days (P=0.056)

Figure 3. All-Cause HCRU: Schizophrenia Cohort (n=855)[a]

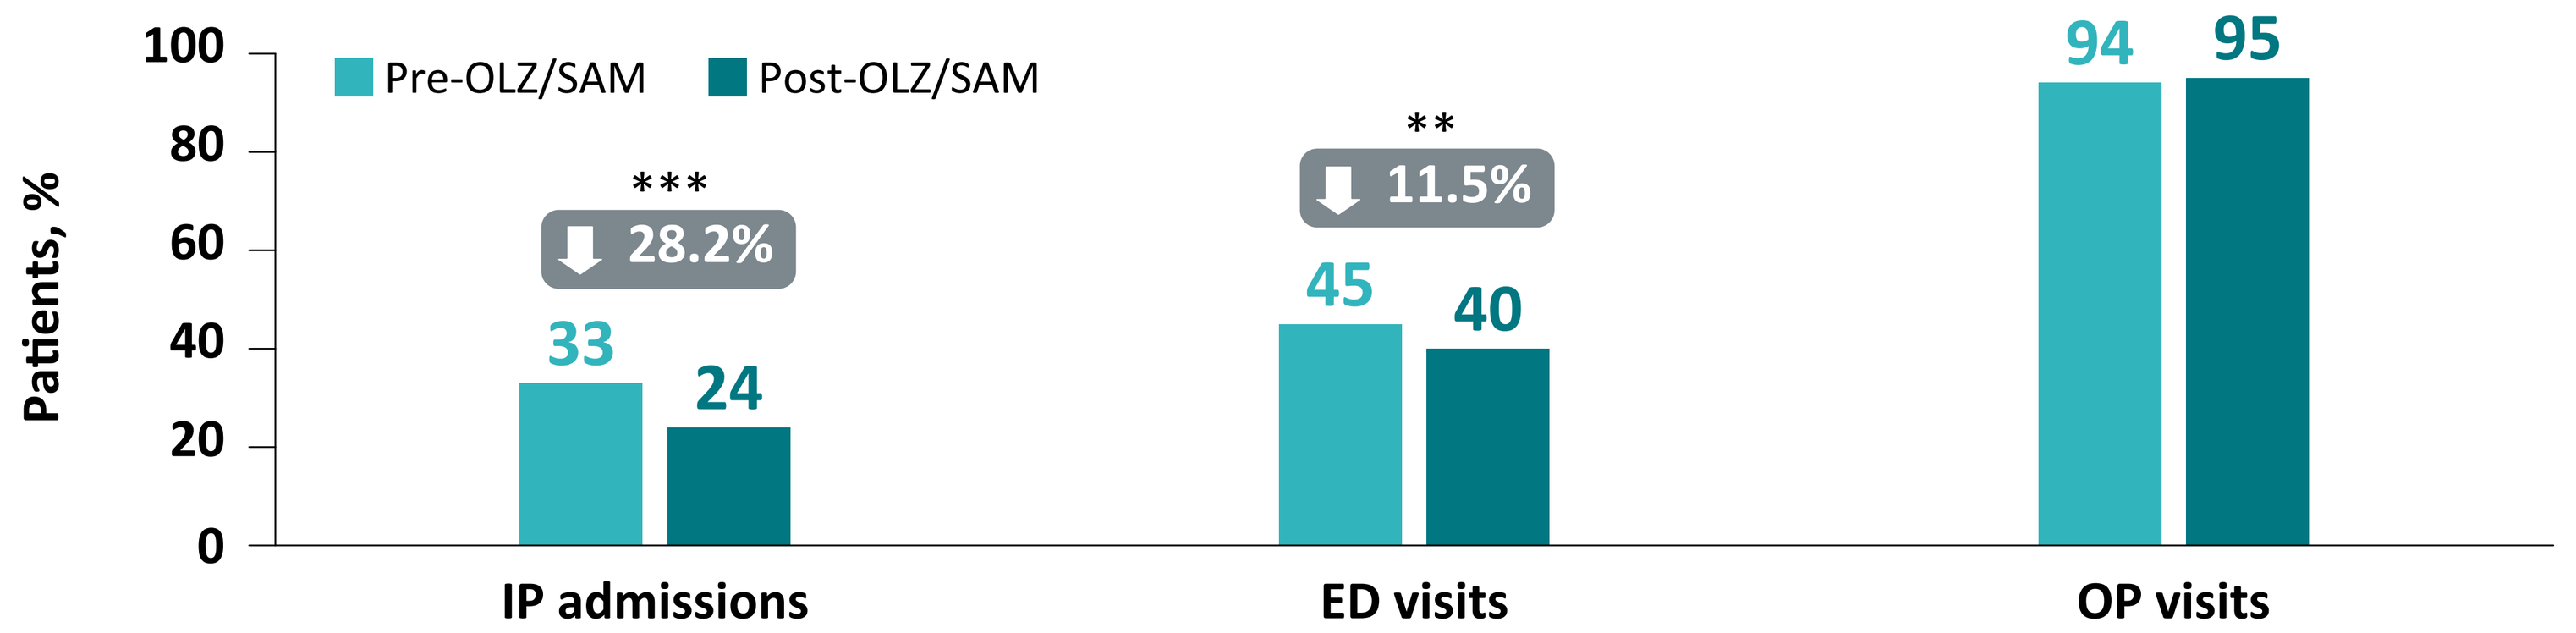
100
80
60
40
20
0
Patients, %
Pre-OLZ/SAM
Post-OLZ/SAM

28.2%
33
24
IP admissions
**
11.5%
45
40
ED visits
94
95
OP visits

P<0.01; *P<0.001.
[a]Gray boxes represent percent change from baseline.
ED, emergency department; HCRU, healthcare resource utilization; IP, inpatient; OLZ/SAM, combination olanzapine and samidorphan; OP, outpatient; SZ, schizophrenia.

Figure 4. Mental Health–Related HCRU: Schizophrenia Cohort (n=855)[a]

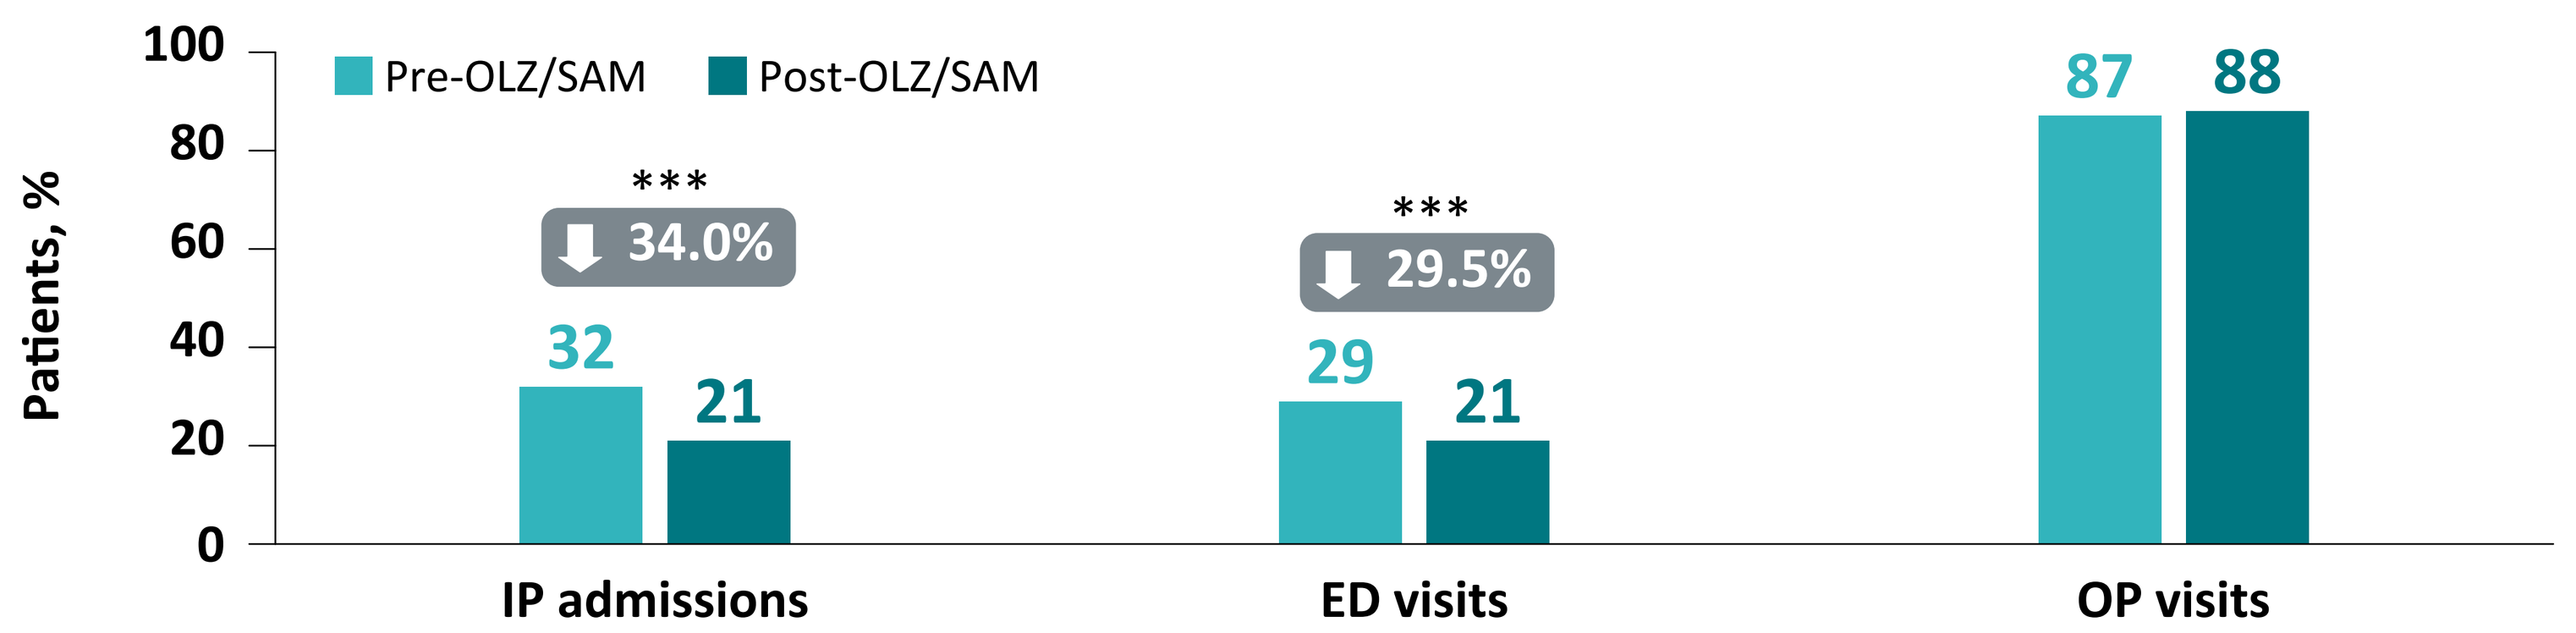
100
80
60
40
20
0
Patients, %
Pre-OLZ/SAM
Post-OLZ/SAM

34.0%
32
21
IP admissions

29.5%
29
21
ED visits
87
88
OP visits

***P<0.001.
[a]Gray boxes represent percent change from baseline.
ED, emergency department; HCRU, healthcare resource utilization; IP, inpatient; OLZ/SAM, combination olanzapine and samidorphan; OP, outpatient; SZ, schizophrenia.

HCRU in BD-I Cohort

- Significant reductions from baseline in the proportions of patients with all-cause (**Figure 5**) and mental health–related (**Figure 6**) IP admissions and ED visits were observed in the 6 months after OLZ/SAM initiation
 - Average all-cause number of days in hospital per BD-I patient decreased from 3.4 to 2.2 days (P=0.011)
 - Average mental health–related number of days in hospital per BD-I patient decreased from 3.3 to 2.0 days (P=0.007)

Figure 5. All-Cause HCRU: Bipolar I Disorder Cohort (n=691)[a]

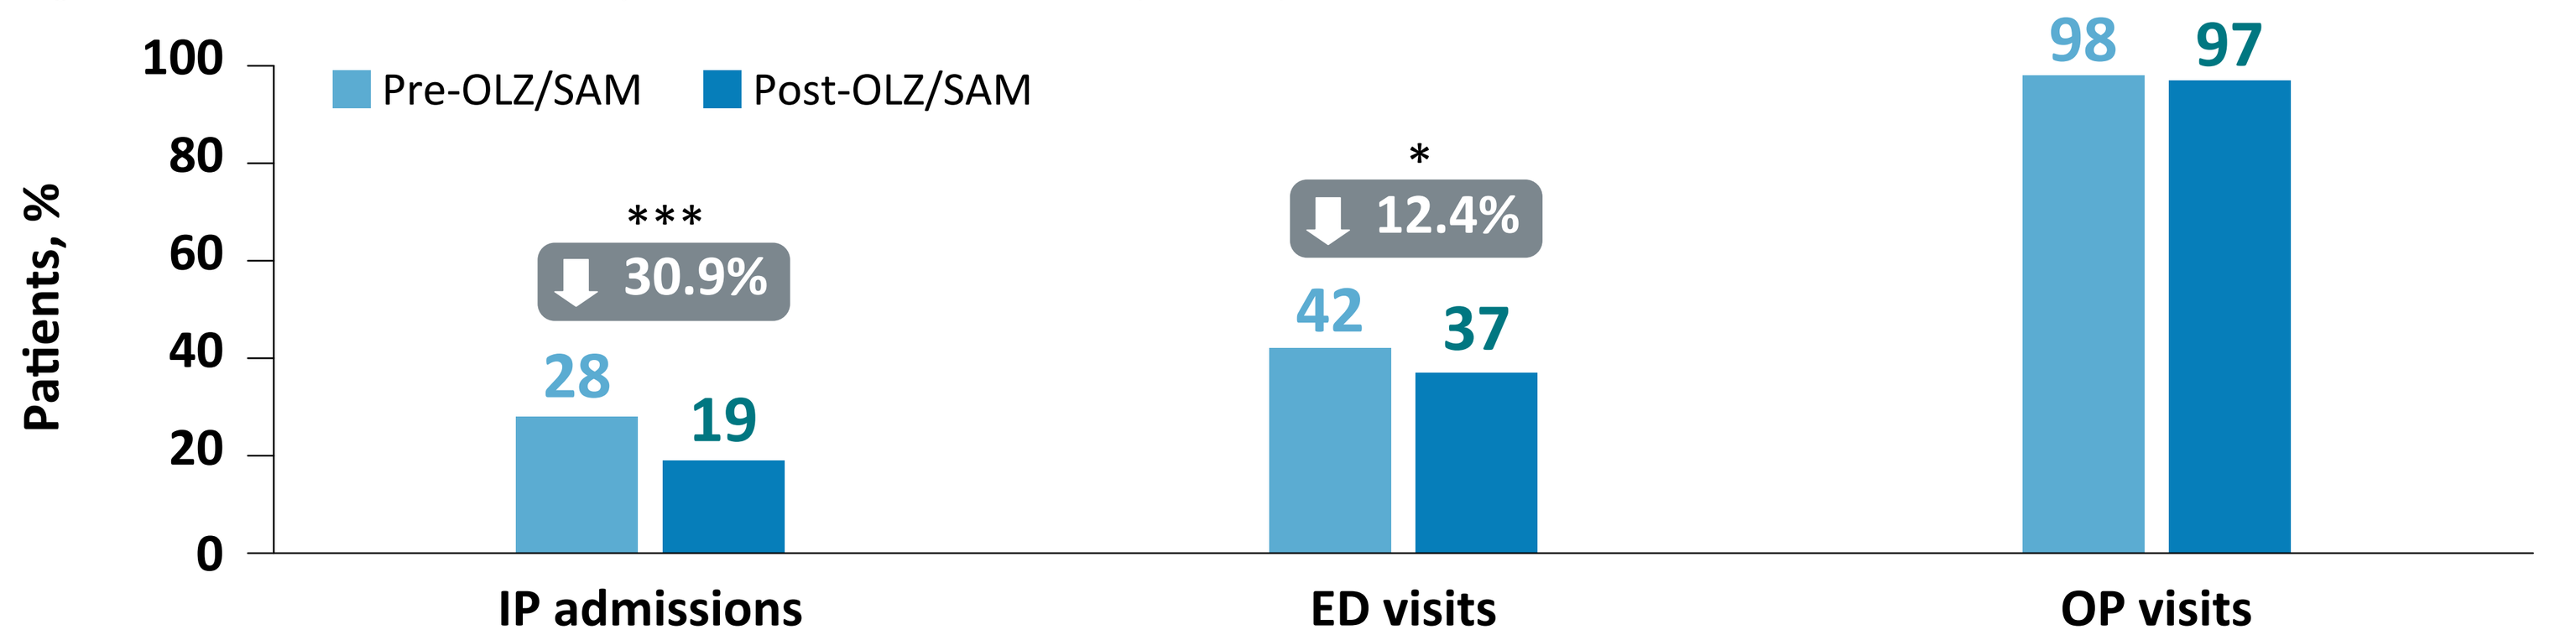
100
80
60
40
20
0
Patients, %
Pre-OLZ/SAM
Post-OLZ/SAM

30.9%
28
19
IP admissions
*
12.4%
42
37
ED visits
98
97
OP visits

*P<0.05; ***P<0.001.
[a]Gray boxes represent percent change from baseline.
BD-I, bipolar I disorder; ED, emergency department; HCRU, healthcare resource utilization; IP, inpatient; OLZ/SAM, combination olanzapine and samidorphan; OP, outpatient.

Figure 6. Mental Health–Related HCRU: Bipolar I Disorder Cohort (n=691)[a]

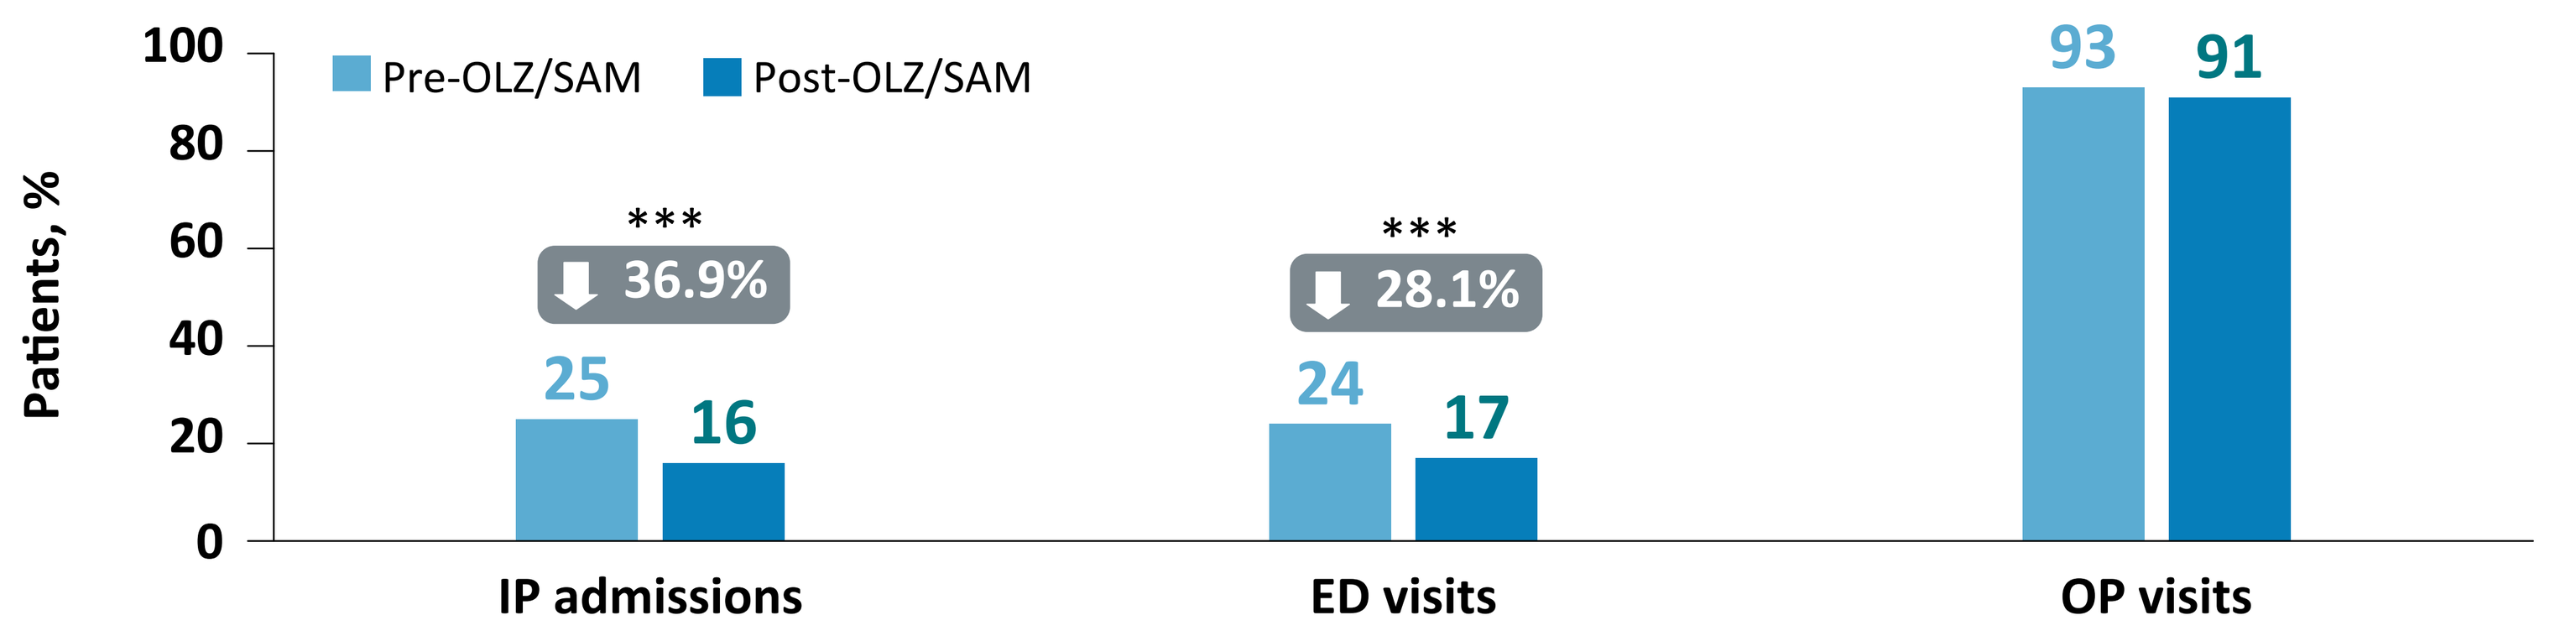
100
80
60
40
20
0
Patients, %
Pre-OLZ/SAM
Post-OLZ/SAM

36.9%
25
16
IP admissions

28.1%
24
17
ED visits
93
91
OP visits

***P<0.001.
[a]Gray boxes represent percent change from baseline.
BD-I, bipolar I disorder; ED, emergency department; HCRU, healthcare resource utilization; IP, inpatient; OLZ/SAM, combination olanzapine and samidorphan; OP, outpatient.

DISCONTINUATION, ADHERENCE, AND PERSISTENCE

Table 3. Discontinuation, Adherence, and Persistence Post-OLZ/SAM Initiation

Follow-up[a] treatment patterns	SZ Cohort (n=855)	BD-I Cohort (n=691)
Discontinuation of index therapy,[b] n (%)	409 (47.84)	389 (56.30)
Medication possession ratio,[c] mean (SD)	0.88 (0.18)	0.87 (0.19)
Proportion of days covered,[d] mean (SD)	0.64 (0.33)	0.57 (0.33)
Days persistent,[e] mean (SD)	121.97 (66.88)	111.09 (67.16)

[a]The follow-up period begins on the index date and ends 6 months after the index date. [b]Defined as a minimum 45-day gap in therapy. [c]Calculated as sum of days' supply of index medication during follow-up divided by number of days in the follow-up period. [d]Calculated as number of days for which medication was available (based on filled prescriptions) divided by the number of days between index date and follow-up period. [e]Measured as the number of days from the index date to the discontinuation date (for patients who discontinued) or from the index date to the end of the follow-up period (for patients who did not discontinue).
BD-I, bipolar I disorder; OLZ/SAM, combination olanzapine and samidorphan; SZ, schizophrenia.

COMPLETER ANALYSIS

- In completers with SZ (n=402), all-cause IP admissions and ED visits were reduced by 50.0% and 28.5%, respectively, relative to baseline; mental health–related IP admissions and ED visits were reduced by 56.6% and 52.6%, respectively, relative to baseline; P<0.001 for each
- In completers with BD-I (n=302), all-cause IP admissions and ED visits decreased by 42.9% and 30.8%, respectively, relative to baseline; mental health–related IP admissions and ED visits decreased by 47.8% and 60.3%, respectively, relative to baseline; P<0.001 for each
- All-cause and mental health–related numbers of days in hospital per patient were significantly reduced from baseline in the SZ cohort (P<0.05) but not in the BD-I cohort

LIMITATIONS

- Results from the insured population studied may not be generalizable to uninsured populations
- A claim for a filled prescription may not indicate that the medication was consumed or taken as prescribed
- The presence of a diagnosis code may not definitively be indicative of disease presence or causality, nor does the presence of a claim indicate disease severity
- Because of the limited follow-up time, HCRU reported may not fully capture the effects of longer-term OLZ/SAM use

CONCLUSIONS

- In this first real-world data assessment of HCRU use in patients with SZ or BD-I, OLZ/SAM initiation was associated with significant reductions in HCRU rates for IP admissions and ED visits
 - The proportions of patients with all-cause and mental health–related IP admissions and ED visits decreased significantly between baseline and follow-up periods in patients with SZ and in those with BD-I
 - Average number of days in hospital per patient was reduced between the baseline and follow-up periods by >1 day in both the SZ and BD-I cohorts; reductions were statistically significant in patients with BD-I for both all-cause and mental health–related admissions
 - Patients remaining on OLZ/SAM for the full 6 months had an even greater reduction in HCRU
- Overall, findings indicate that the use of OLZ/SAM may result in clinically meaningful reductions in patient and provider burden, as evidenced by changes in IP care and ED visits

REFERENCES

1. American Psychiatric Association. The American Psychiatric Association Practice Guideline for the Treatment of Patients With Schizophrenia. Washington, DC: American Psychiatric Association; 2021. 2. Yatham LN, et al. *Bipolar Disord*. 2018;20(2):97-170. DOI: https://doi.org/10.1111/bdi.12609. 3. Bessonova L, et al. *BMC Psychiatry*. 2020;20(1):354. DOI: https://doi.org/10.1186/s12888-020-02767-x. 4. Law MR, et al. *J Clin Psychiatry*. 2008;69(1):47-53. DOI: https://doi.org/10.4088/jcp.v69n0107. 5. Perkins AJ, et al. *BMC Psychiatry*. 2023;23(1):64. DOI: https://doi.org/10.1186/s12888-023-04558-6. 6. Leucht S, et al. *Lancet*. 2013;382(9896):951-62. DOI: https://doi.org/10.1016/s0140-6736(13)60733-3. 7. De Hert M, et al. *Nat Rev Endocrinol*. 2012;8(2):114-26. DOI: https://doi.org/10.1038/nrendo.2011.156. 8. Martin WF, et al. *Am J Psychiatry*. 2019;176(6):457-67. DOI: https://doi.org/10.1176/appi.ajp.2018.18030280. 9. Correll CU, et al. *Am J Psychiatry*. 2020;177(12):1168-78. DOI: https://doi.org/10.1176/appi.ajp.2020.19121279. 10. Kahn RS, et al. *J Clin Psychiatry*. 2023;84(3):22m14674. DOI: https://doi.org/10.4088/JCP.22m14674.

Disclosure information, acknowledgments, and copies of this poster can be obtained through this QR (Quick Response) code. These materials are for personal use only and may not be reproduced without permission of Alkermes. For permission, contact USMedInfo@Alkermes.com

Supported by funding from Alkermes, Inc.

Healthcare Resource Utilization Following 6 Months of Treatment With Olanzapine/Samidorphan: Real-World Assessment of Patients With Schizophrenia or Bipolar I Disorder

Andrew Cutler,[1,2] Hemangi R. Panchmatia,[3] Alejandro G. Hughes,[4] Noah S. Webb,[4] Michael J. Doane,[3] Rakesh Jain[5]

[1]SUNY Upstate Medical University, Syracuse, NY; [2]Neuroscience Education Institute, Lakewood Ranch, FL; [3]Alkermes, Inc., Waltham, MA; [4]Optum, Inc., Eden Prairie, MN; [5]Department of Psychiatry, Texas Tech University School of Medicine-Permian Basin, Midland, TX

AUTHOR DISCLOSURES

RJ has served as a consultant for AbbVie, Acadia, Adamas, Alfasigma, Alkermes, Almatica, Axsome, Biogen, Boehringer Ingelheim, Cingulate Therapeutics, Corium, Eisai, Evidera, Impel, Janssen, Lilly, Lundbeck, Merck, Neos Therapeutics, Neurocrine Biosciences, Osmotica, Otsuka, Pamlab, Pfizer, Sage Therapeutics, Shire, Sunovion, Supernus, Takeda, Teva, Transcend Therapeutics, and Viatris; has received speaker/promotional honoraria from AbbVie, Alkermes, Almatica, Axsome, Corium, Eisai, Intra-Cellular Therapies, Ironshore Pharmaceuticals, Janssen, Lilly, Lundbeck, Merck, Neos Therapeutics, Otsuka, Pamlab, Pfizer, Shire, Sunovion, Takeda, Tris Pharmaceuticals, and Viatris; has served on advisory boards for Adamas, Alkermes, Corium, Eisai, Janssen, Lilly, Lundbeck, Merck, Neos Therapeutics, Neurocrine Biosciences, Otsuka, Pamlab, Pfizer, Sage Therapeutics, Shire, Sunovion, Supernus, Takeda, and Teva; and has received research funding from AbbVie, Lilly, Lundbeck, Otsuka, Pfizer, Shire, and Takeda. **HRP** and **MJD** are or were employees of Alkermes, Inc., and may own stock/options in the company. **AGH** and **NSW** are employees of Optum, Inc., a health services innovation company that received funding from Alkermes, Inc., to conduct this study and analyze the data used for this publication. **AJC** has served on advisory boards for Alkermes, Acadia, Biogen, BioXcel, Intra-Cellular Therapies, and Teva and on speakers bureaus for Acadia, Axsome, BioXcel, Intra-Cellular Therapies, Neurocrine, and Teva.

ACKNOWLEDGMENTS

This study was sponsored by Alkermes, Inc. (Waltham, MA, USA). Medical writing and editorial support were provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Alkermes, Inc.

Supported by funding from Alkermes, Inc.