PCR46

Preferences for Characteristics of Oral Antipsychotic Treatments: Survey Results of Patients Living With Schizophrenia or Bipolar I Disorder

Michael J. Doane,[1] Marco Boeri,[2,3] Caroline Vass,[4,5] Cooper Bussberg,[6] Hemangi R. Panchmatia,[1] Sabina Gasper,[1] Leslie Citrome,[7] Martha Sajatovic[8]

[1]Alkermes, Inc., Waltham, MA, USA; [2]Health Preference Assessment, RTI Health Solutions, Belfast, Northern Ireland, UK; [3]Queen's University Belfast, Belfast, Northern Ireland, UK; [4]University of Manchester, Manchester, England, UK; [5]Health Preference Assessment, RTI Health Solutions, Manchester, England, UK; [6]Health Preference Assessment, RTI Health Solutions, Research Triangle Park, NC, USA; [7]New York Medical College, Valhalla, NY, USA; [8]University Hospitals of Cleveland Medical Center, Case Western Reserve University School of Medicine, Cleveland, OH, USA

INTRODUCTION

- Antipsychotic medications are effective in managing symptoms of schizophrenia (SZ) and bipolar I disorder (BD-I)[1,2] but are associated with various side effects (eg, weight gain, sexual dysfunction)[1]
- Previous research has suggested that patients may be willing to make tradeoffs between clinical improvement and side effect profiles when choosing among antipsychotic treatments[3,4]

OBJECTIVE

- This study assessed patient preferences for characteristics associated with oral antipsychotics and further explored potential tradeoffs that patients may make between efficacy and side effects

METHODS

Study Design

- A cross-sectional online survey designed to collect preference data using a discrete-choice experiment (DCE)
- The DCE consisted of a series of choices between pairs of hypothetical oral antipsychotic treatments based on 5 characteristics: treatment efficacy (ie, symptom improvement), weight gain over 6 months, sexual dysfunction, sedation, and akathisia (ie, restlessness caused by antipsychotic medication)
- The DCE was pretested to ensure its comprehension and understanding among people living with SZ (n=15) or BD-I (n=15)
- The final survey was administered to US adults aged ≥18 to 64 years with a self-reported diagnosis of SZ or BD-I

Outcomes

- The primary outcome of the survey was respondents' preferences for each level assessed across the 5 treatment characteristics
 - Preference coefficient estimates for each level were derived from random-parameter logit models
 - Within each characteristic, higher preference coefficient estimates for a given level are associated with a greater preference
- Secondary outcomes included the relative importance of individual treatment characteristics, as well as the willingness to accept treatment-related side effects of weight gain and sedation as a tradeoff for improvement in antipsychotic efficacy
 - The conditional relative importance (CRI) of each treatment characteristic was calculated by subtracting the preference coefficient estimate of the least-preferred level from that of the most-preferred level
 - Preference coefficient estimates were also used to calculate the maximum acceptable weight gain and the maximum acceptable risk of sedation that respondents were willing to accept for symptom improvement

RESULTS

TABLE 1. Demographic Characteristics

Parameter	SZ Cohort (n=144)	BD-I Cohort (n=152)
Female, n (%)	72 (50.0)	106 (69.7)
White, n (%)	100 (69.4)	118 (77.6)
Age, mean (SD), years	41.0 (10.1)	40.0 (10.7)
Age at diagnosis, mean (SD), years	30.0 (10.5)	25.0 (9.1)
≤5 years since diagnosis, n (%)	64 (44.4)	34 (22.4)
FGA treatment exposure, n (%)		
Currently taking	34 (23.6)	9 (5.9)
Taken in the past	64 (44.4)	47 (30.9)
SGA treatment exposure, n (%)		
Currently taking	91 (63.2)	59 (38.8)
Taken in the past	71 (49.3)	97 (63.8)
Symptom severity in past week, n (%)		
No symptoms	6 (4.2)	14 (9.2)
Mild	34 (23.6)	35 (23.0)
Moderate	65 (45.1)	57 (37.5)
Severe	39 (27.1)	46 (30.3)
BMI, mean (SD), kg/m^2	32.0 (8.6)	32.6 (9.1)
Overweight,[a] n (%)	43 (29.9)	30 (19.7)
Obese,[b] n (%)	75 (52.1)	84 (55.3)
Experienced antipsychotic side effects, n (%)		
Weight gain	123 (85.4)	126 (82.9)
Sedation	118 (81.9)	142 (93.4)
Akathisia	102 (70.8)	110 (72.4)
Sexual dysfunction	92 (63.9)	105 (69.1)

[a]Overweight is defined as BMI >25.0 to <30 kg/m^2. [b]Obese is defined as BMI ≥30.0 kg/m^2.
BD-I, bipolar I disorder; BMI, body mass index; FGA, first-generation antipsychotic; SGA, second-generation antipsychotic; SZ, schizophrenia.

FIGURE 1. Preferences for Characteristics of Oral Antipsychotics for Respondents With SZ or BD-I[a]

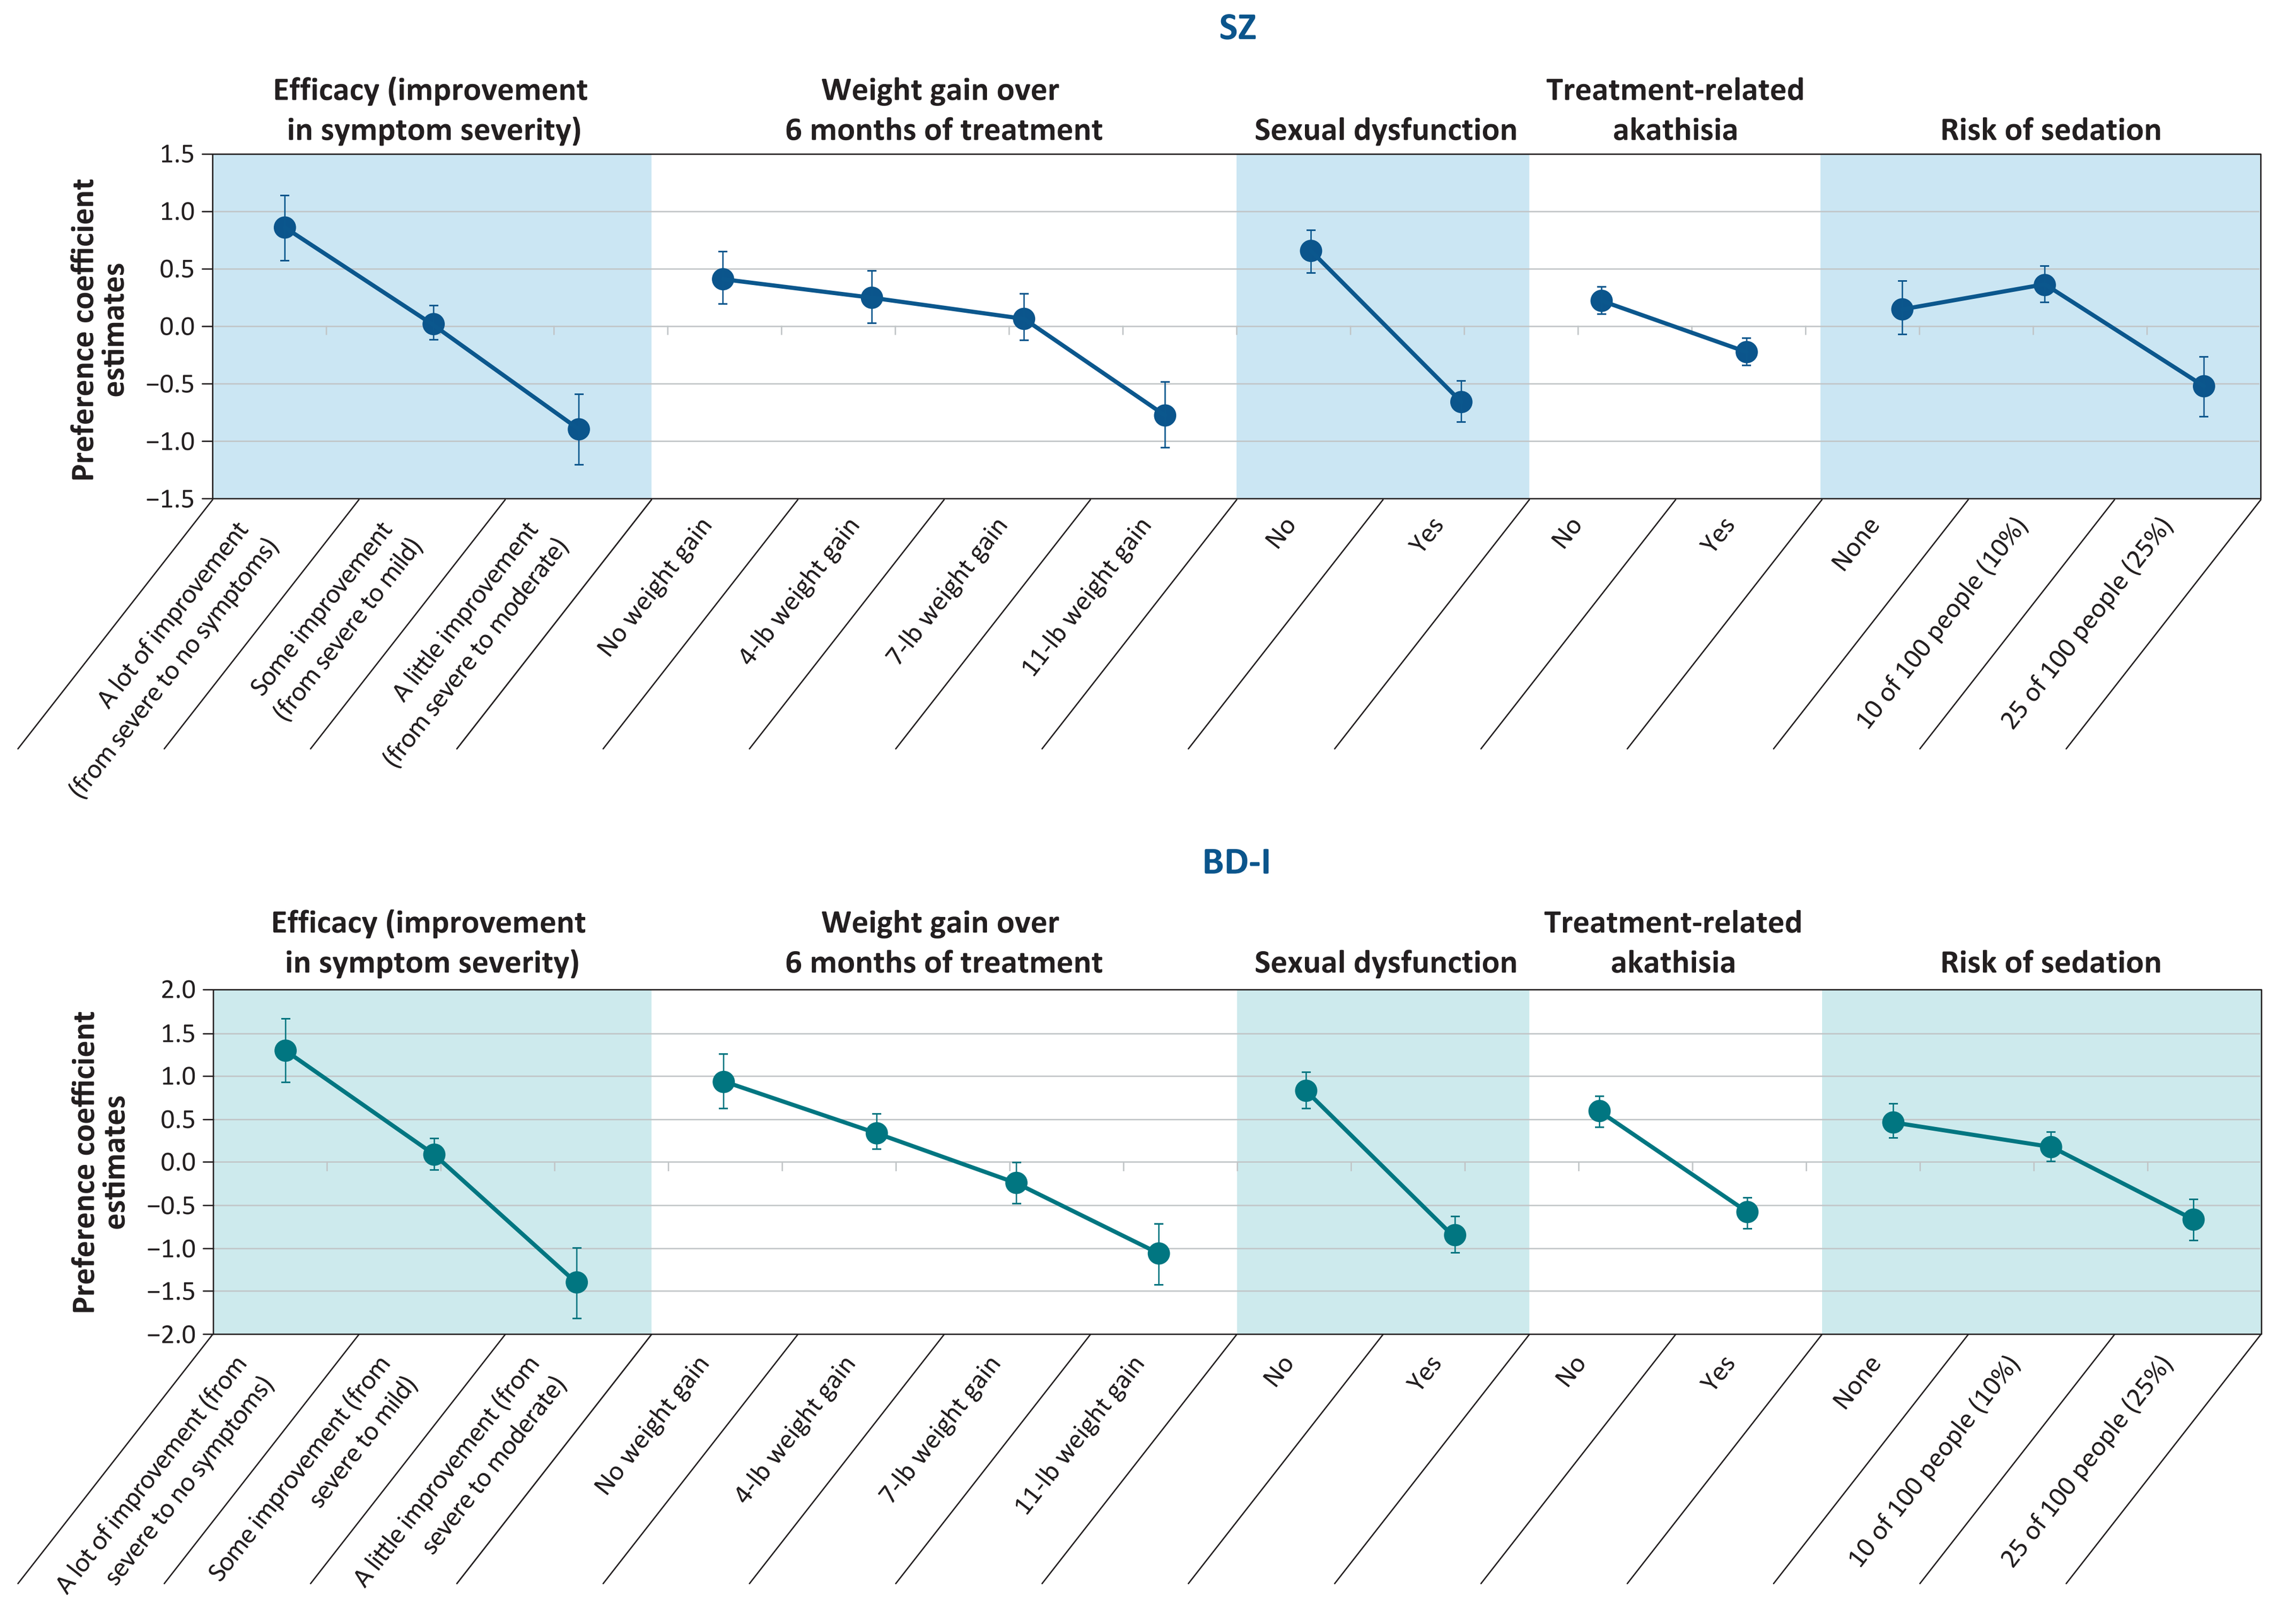
SZ
BD-I
Efficacy (improvement in symptom severity)
Weight gain over 6 months of treatment
Sexual dysfunction
Treatment-related akathisia
Risk of sedation
Preference coefficient estimates
A lot of improvement (from severe to no symptoms)
Some improvement (from severe to mild)
A little improvement (from severe to moderate)
No weight gain
4-lb weight gain
7-lb weight gain
11-lb weight gain
No
Yes
None
10 of 100 people (10%)
25 of 100 people (25%)

[a]Preference coefficient estimates are presented along with their 95% CIs, with higher estimates for a given level associated with a greater preference for that level. The vertical distance between any 2 levels of a characteristic represents the change in utility; larger differences indicate that respondents viewed the change as having a relatively greater effect on overall utility. Characteristics are presented in the order in which they appeared in the DCE questions.
BD-I, bipolar I disorder; DCE, discrete-choice experiment; SZ, schizophrenia.

FIGURE 2. Relative Importance of Oral Antipsychotic Treatment Characteristics for Respondents With SZ or BD-I[a]

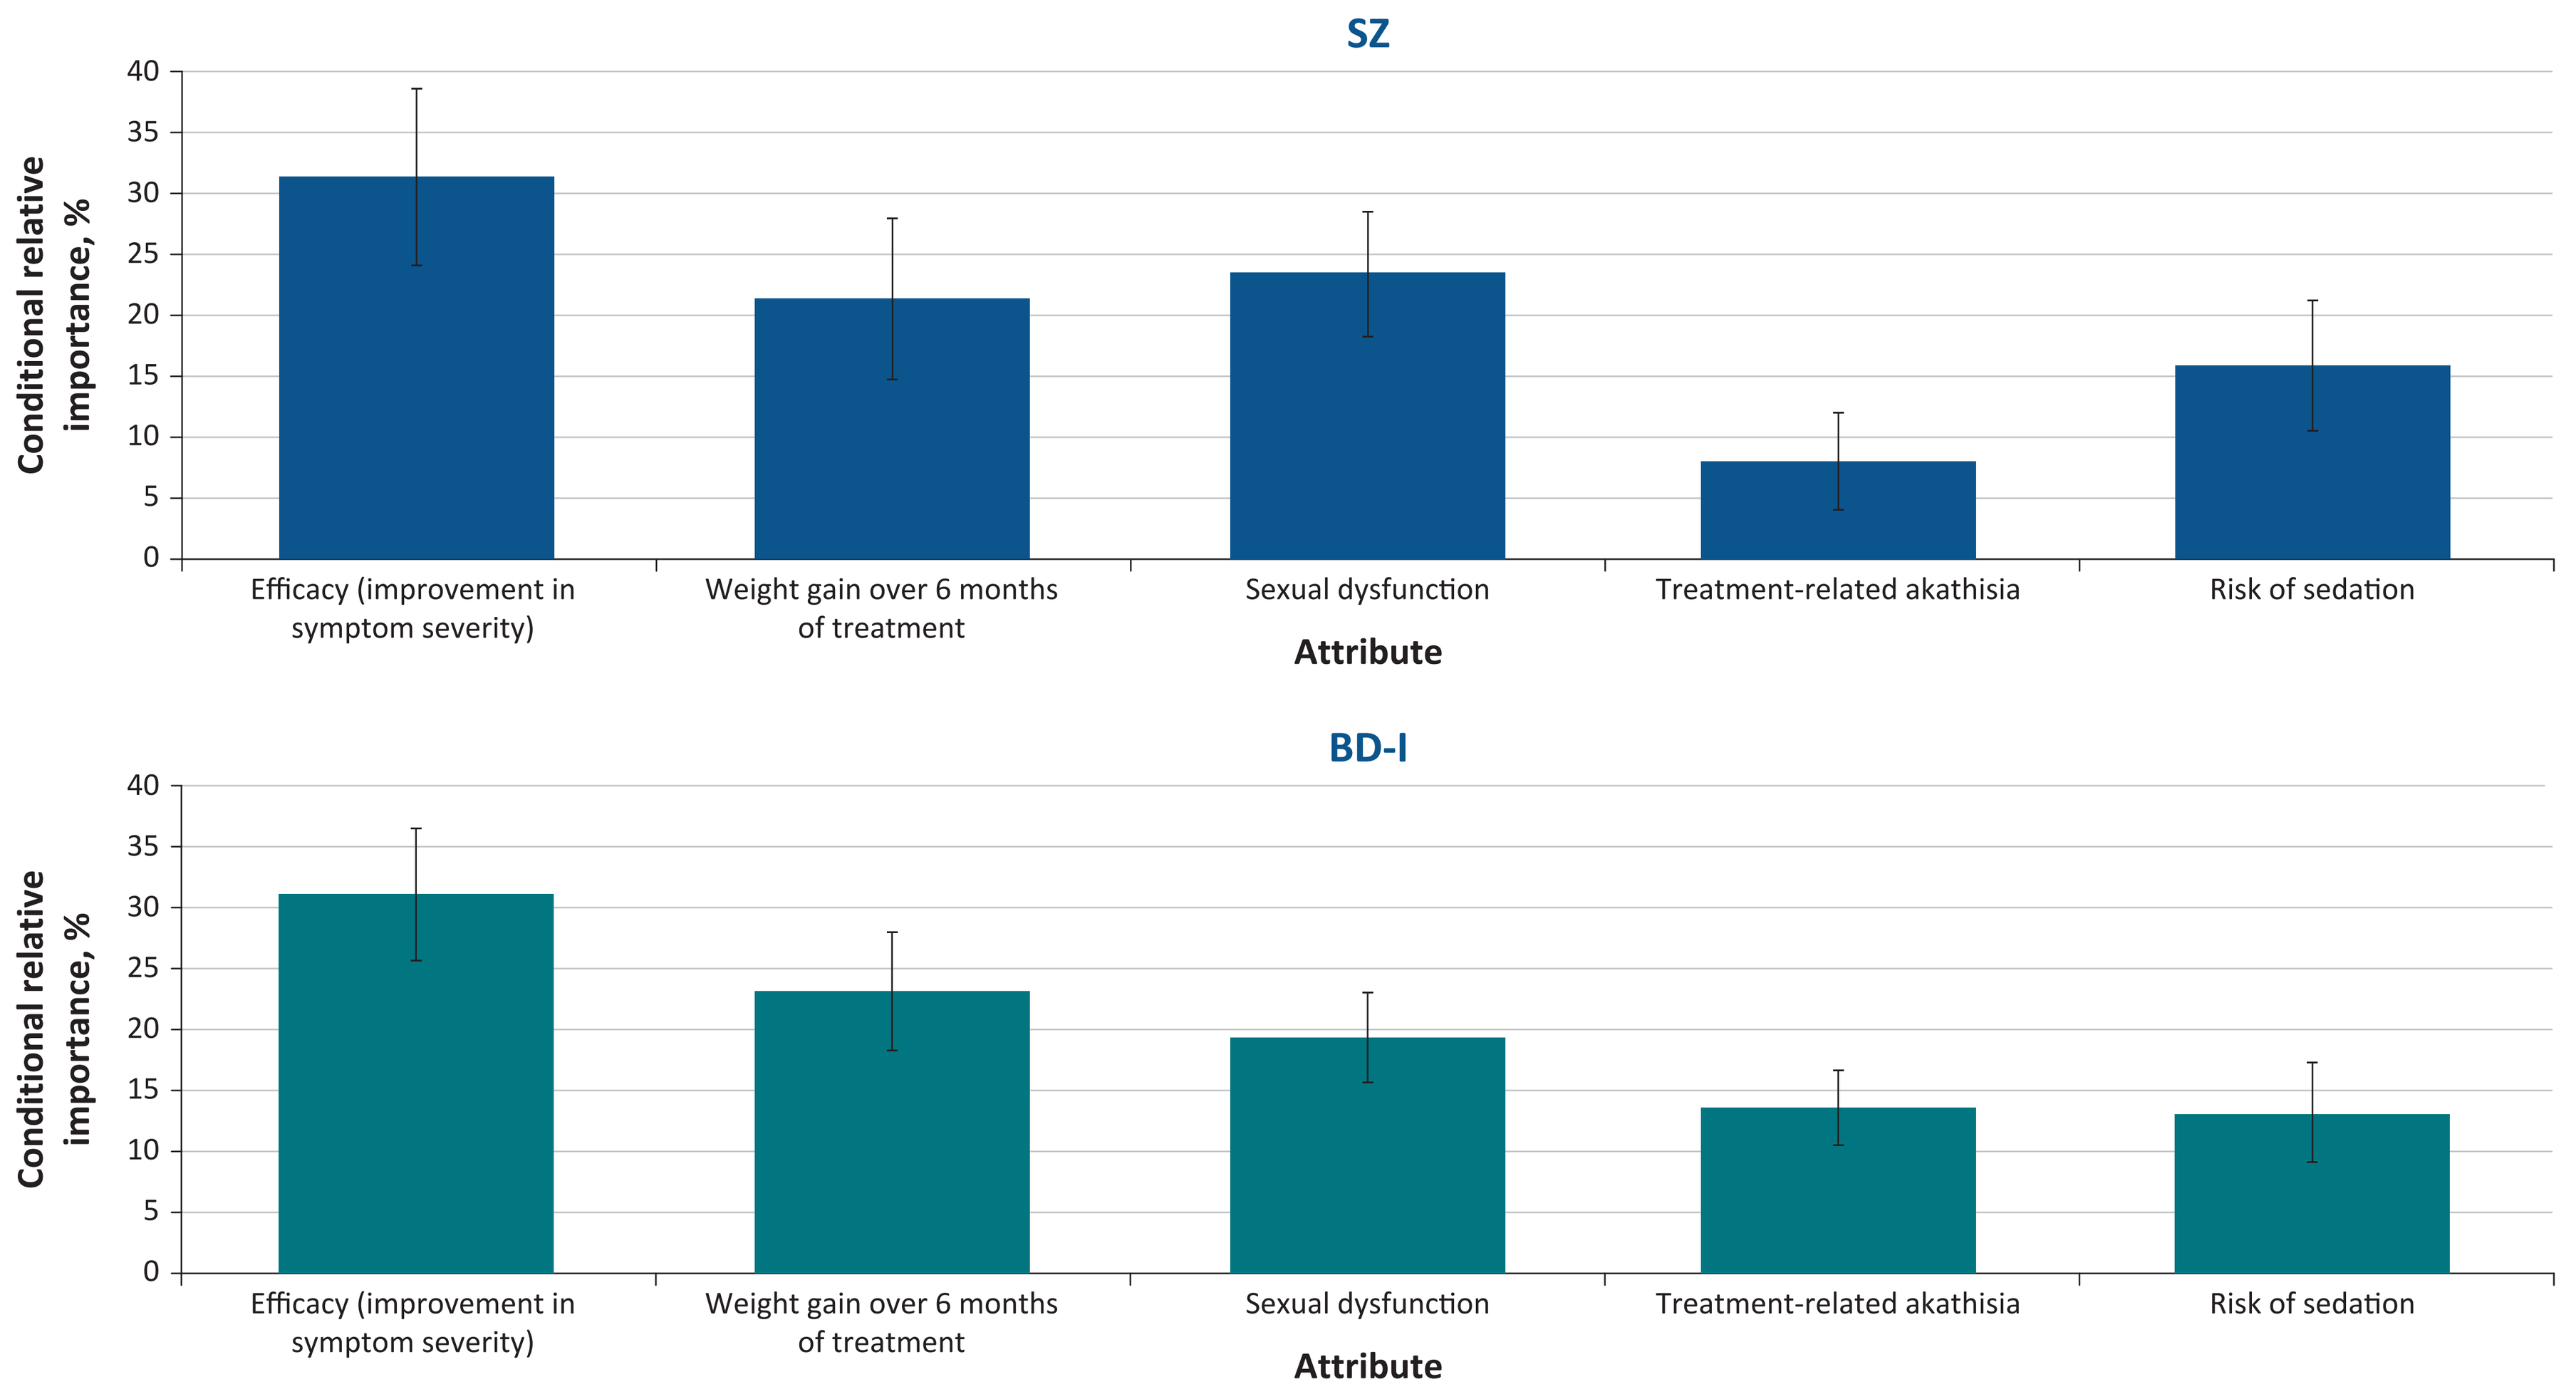
SZ
BD-I
Conditional relative importance, %
Efficacy (improvement in symptom severity)
Weight gain over 6 months of treatment
Sexual dysfunction
Treatment-related akathisia
Risk of sedation
Attribute

[a]Conditional relative importance (CRI) is interpreted as the proportion of utility gained by improving each characteristic from the least-preferred to the most-preferred level, relative to the maximum utility gained by improving all characteristics. Each CRI was calculated by subtracting the preference coefficient estimate of the least-preferred level from that of the most-preferred level. Differences were summed across characteristics and rescaled to 100. Each CRI is presented as a percentage of this total along with its 95% CI. Characteristics are presented in the order in which they appeared in the DCE questions.
BD-I, bipolar I disorder; DCE, discrete-choice experiment; SZ, schizophrenia.

- Symptom improvement was the most important treatment characteristic endorsed by respondents with SZ or BD-I (**Figure 1**)
- Weight gain and sexual dysfunction were the side effects that respondents across both cohorts most wanted to avoid (**Figure 2**)
- Respondents with SZ wanted to avoid antipsychotic treatments associated with 11 lb of weight gain over 6 months vs those associated with 0, 4, or 7 lb of weight gain over 6 months
- Respondents with BD-I showed a more linear pattern regarding weight gain avoidance; avoiding weight gain of 11 vs 7 lb over 6 months was more important than avoiding weight gain of 7 vs 4 lb and of 4 vs 0 lb over 6 months
- To achieve 1 incremental step of disease severity improvement (ie, "some" to "a lot" of improvement or "a little" to "some" improvement over 6 months of treatment):
 - Respondents with SZ were willing to accept a weight increase of 9.3 or 9.8 lb, respectively
 - Patients with BD-I were willing to accept a weight increase of 7.1 or 8.5 lb, respectively
- For 2 incremental steps of disease severity improvement (eg, "a little" to "a lot" of improvement), respondents from both cohorts were willing to accept a weight gain of >11 lb over 6 months
- Respondents with SZ or BD-I were willing to accept a >25% risk of sedation for any level of symptom improvement

LIMITATIONS

- SZ or BD-I diagnosis was self-reported by respondents and not confirmed by clinician assessment
- The DCE included a maximum acceptable risk of sedation of 25%; thus, the relative importance of this side effect may be underestimated
- This sample may not be representative of the population of people living with SZ or BD-I

CONCLUSIONS

- Treatment efficacy was endorsed as the most important attribute of oral antipsychotics among respondents with SZ or BD-I
 - Weight gain and sexual dysfunction were the 2 side effects that respondents most wanted to avoid
 - This is consistent with previous studies reporting that weight gain and sexual dysfunction are among the most bothersome side effects of antipsychotic treatments[3,4]
- Respondents with SZ or BD-I were willing to accept some degree of weight gain as a side effect in exchange for better antipsychotic medication efficacy
 - Although weight gain is associated with medical comorbidities,[5] the degree to which patients are willing to accept the risk of weight gain in return for better symptom control may help inform treatment decisions
- As oral antipsychotics have different efficacy and side effect profiles, it is important to understand the features that patients value in a treatment and how they balance benefits and risks when choosing among treatments

REFERENCES

1. Patel KR, et al. *P T.* 2014;39:638-45.
2. Goldberg JF, Citrome L. *Postgrad Med.* 2005;117:25-6, 29-32, 35-6.
3. Doane MJ, et al. *Patient Prefer Adherence.* 2020;14:2043-54.
4. Bessonova L, et al. *BMC Psychiatry.* 2020;20:354.
5. Zheng Y, et al. *JAMA.* 2017;318:255-69.

AUTHOR DISCLOSURES

MJD, HRP, and **SG** are or were employees of Alkermes, Inc., and may own stock/options in the company.

MB, CV, and **CB** are employees of RTI Health Solutions, a not-for-profit research institute that received funding from Alkermes, Inc., to conduct this study and collect the data used for this publication.

LC consulted with Alkermes on this research and has served as a consultant for AbbVie/Allergan, Acadia, Adamas, Angelini, Astellas, Avanir, Axsome, BioXcel, Boehringer Ingelheim, Cadent Therapeutics, Cerevel, Clinilabs, COMPASS, Eisai, Enteris BioPharma, HLS Therapeutics, Idorsia, Impel, INmune Bio, Intra-Cellular Therapies, Janssen, Karuna, Lundbeck, Lyndra, MedAvante-ProPhase, Marvin, Merck, Mitsubishi-Tanabe Pharma, Neurelis, Neurocrine, Novartis, Noven, Otsuka, Ovid, Praxis, Recordati, Relmada, Reviva, Sage, Sunovion, Supernus, Teva, and University of Arizona and performed one-off ad hoc consulting for individuals/entities conducting marketing, commercial, or scientific scoping research; has served as a speaker for AbbVie/Allergan, Acadia, Alkermes, Angelini, Axsome, BioXcel, Eisai, Idorsia, Intra-Cellular Therapies, Janssen, Lundbeck, Neurocrine, Noven, Otsuka, Recordati, Sage, Sunovion, Takeda, Teva, and CME activities organized by medical education companies, such as Medscape, NACCME, NEI, Vindico, and universities and professional organizations/societies; owns stocks (small number of shares of common stock) in Bristol-Myers Squibb, Eli Lilly, J&J, Merck, and Pfizer (purchased >10 years ago) and stock options in Reviva; and receives royalties/publishing income from Taylor & Francis (Editor-in-Chief, *Current Medical Research and Opinion*, 2022-date), Wiley (Editor-in-Chief, *International Journal of Clinical Practice*, through end of 2019), UpToDate (reviewer), Springer Healthcare (book), and Elsevier (Topic Editor, Psychiatry, *Clinical Therapeutics*).

MS consulted with Alkermes on this research and has served as a consultant to Janssen, Lundbeck, Neurelis, Otsuka, and Teva; has received research grants (within 3 years) from Centers for Disease Control and Prevention, International Society for Bipolar Disorders, National Institutes of Health, and Patient-Centered Outcomes Research Institute; has received royalties from Johns Hopkins University Press, Oxford Press, Springer Press, and UpToDate; and has prepared CME activities for the American Academy of Child and Adolescent Psychiatry, American Epilepsy Society, American Physician Institute (CMEtoGO), Clinical Care Options, Neurocrine, and Psychopharmacology Institute.

ACKNOWLEDGMENTS

This study was sponsored by Alkermes, Inc. Medical writing and editorial support were provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Alkermes, Inc.

Copies of this poster obtained through this QR (Quick Response) code are for personal use only and may not be reproduced without permission of Alkermes. For permission, contact USMedInfo@Alkermes.com

Supported by funding from Alkermes, Inc.